Poster 875-P

Baseline characteristics for FINE-ONE: A randomized phase III trial assessing finerenone in people with chronic kidney disease and type 1 diabetes

Hiddo J. L. Heerspink,[1] Andreas L. Birkenfeld,[2–4] David Z. I. Cherney,[5] Helen M. Colhoun,[6] Linong Ji,[7] Chantal Mathieu,[8] Per-Henrik Groop,[9,10] Richard E. Pratley,[11] Sylvia E. Rosas,[12] Peter Rossing,[13,14] Jay S. Skyler,[15] Katherine R. Tuttle,[16] Robert Lawatscheck,[17] Markus F. Scheerer*,[18] Julie Russell,[19] Janet B. McGill[20]

[1]Clinical Pharmacy and Pharmacology, University of Groningen University Medical Center Groningen, Groningen, Netherlands; [2]Institute for Diabetes Research and Metabolic Diseases of the Helmholtz Center Munich at the University of Tübingen, Tübingen, Germany; [3]German Center for Diabetes Research (DZD), Neuherberg, Germany; [4]Department of Internal Medicine, Division of Endocrinology, Diabetology and Nephrology, Eberhard Karls University Tübingen, Tübingen, Germany; [5]Division of Nephrology, University Health Network, Toronto General Hospital, University of Toronto, Toronto, Ontario, Canada; [6]Institute of Genetics and Cancer, College of Medicine and Veterinary Medicine, University of Edinburgh, Edinburgh, UK; [7]Departments of Endocrinology and Metabolism, Peking University People's Hospital, Peking University Diabetes Center, Beijing, China; [8]Clinical and Experimental Endocrinology, Katholieke Universiteit Leuven, Leuven, Belgium; [9]Department of Nephrology, Helsinki University Hospital and Helsinki University, Helsinki, Finland; [10]Folkhälsan Research Center, Biomedicum Helsinki, Helsinki, Finland; [11]AdventHealth Translational Research Institute, Orlando, FL, USA; [12]Kidney and Hypertension Unit, Joslin Diabetes Center, Harvard Medical School, Boston, MA, USA; [13]Steno Diabetes Center Copenhagen, Herlev, Denmark; [14]Department of Clinical Medicine, University of Copenhagen, Copenhagen, Denmark; [15]Department of Medicine, University of Miami, Diabetes Research Institute, University of Miami Miller School of Medicine, Miami, FL, USA; [16]Providence Inland Northwest Health, University of Washington School of Medicine, Spokane, WA, USA; [17]Cardiology and Nephrology Clinical Research, Bayer AG, Research & Development, Pharmaceuticals, Clinical Development, Berlin, Germany; [18]Medical Affairs & Pharmacovigilance, Pharmaceuticals, TA CardioRenal & Heart Disease, Bayer AG, Berlin, Germany; [19]Bayer PLC, Reading, UK; [20]Division of Endocrinology, Metabolism & Lipid Research, Washington University in St. Louis, School of Medicine, St. Louis, MO, USA

*Institution at time of data analysis

Corresponding author: Hiddo J. L. Heerspink, h.j.lambers.heerspink@umcg.nl

Background

- Chronic kidney disease (CKD) develops in approximately 1 in 3 people with type 1 diabetes (T1D)[1] and leads to high risks of kidney failure, dialysis, and mortality[2,3]
 - Among people with T1D, women, older adults, and Asian and Black populations are disproportionately affected by CKD[1]
 - Identification of people who may experience significant decline in kidney function, along with timely intervention, is key to preventing complications[4]
- To reduce the risk of CKD progression in people with CKD and T1D, current American Diabetes Association guidelines recommend optimization of glucose control and renin–angiotensin system blockade with an angiotensin-converting enzyme inhibitor (ACEi) or angiotensin-receptor blocker (ARB) for those with hypertension, as well as lifestyle modification[3]
 - ACEis and ARBs have been available since the 1990s, but no new therapies have been approved for this population since then[5]
 - Therapies to reduce adverse kidney outcomes in people with T1D are urgently needed
- The nonsteroidal mineralocorticoid receptor antagonist finerenone significantly reduced adverse kidney and heart outcomes compared with placebo and was well tolerated among people with CKD and type 2 diabetes receiving optimized doses of ACEi or ARB therapy in FIDELITY[6] (a prespecified pooled analysis of the FIDELIO-DKD and FIGARO-DKD trials)[7,8]
 - Finerenone reduced the composite kidney outcome by 23% compared with placebo and reduced albuminuria, urine albumin-to-creatinine ratio (UACR), by 32% from baseline to Month 4[6]

Aims

- FINE-ONE (NCT05901831) is a 7.5-month clinical trial to assess the efficacy and safety of finerenone versus placebo in people with CKD and T1D
 - The primary efficacy outcome is the relative change in UACR from baseline over 6 months
 - For regulatory approval, UACR measurement will be used as a bridging biomarker to translate evidence from people with CKD and T2D in FIDELITY to the FINE-ONE population[5]
- Baseline characteristics of the FINE-ONE population are presented here

Methods

Study design and patient population

- FINE-ONE is an ongoing, global, phase III trial that randomized 242 participants with CKD and T1D 1:1 to finerenone (10 or 20 mg once daily) or placebo
 - The starting dose of finerenone depends on each participant's estimated glomerular filtration rate (eGFR) level: a lower dose of 10 mg once daily if eGFR is 25–<60 mL/min/1.73 m^2, or the higher (target) dose of 20 mg once daily if eGFR is ≥60 mL/min/1.73 m^2
 - Finerenone will be uptitrated to the target dose of 20 mg after 1 month if the serum/plasma potassium concentration ($[K^+]$) is ≤4.8 mmol/L and the eGFR decrease is <30% compared with the value measured at the prior visit
- The study design, outcomes, and inclusion/exclusion criteria for FINE-ONE are shown in **Figure 1**
 - Participants with CKD (defined as UACR 200–<5000 mg/g and eGFR 25–<90 mL/min/1.73 m^2) and T1D who had glycated hemoglobin (HbA1c) <10%, serum $[K^+]$ ≤4.8 mmol/L, and who were on a stable dose of ACEi/ARB were included
 - The primary efficacy outcome is the change in UACR from baseline (ratio to baseline) over 6 months

Figure 1. FINE-ONE study details

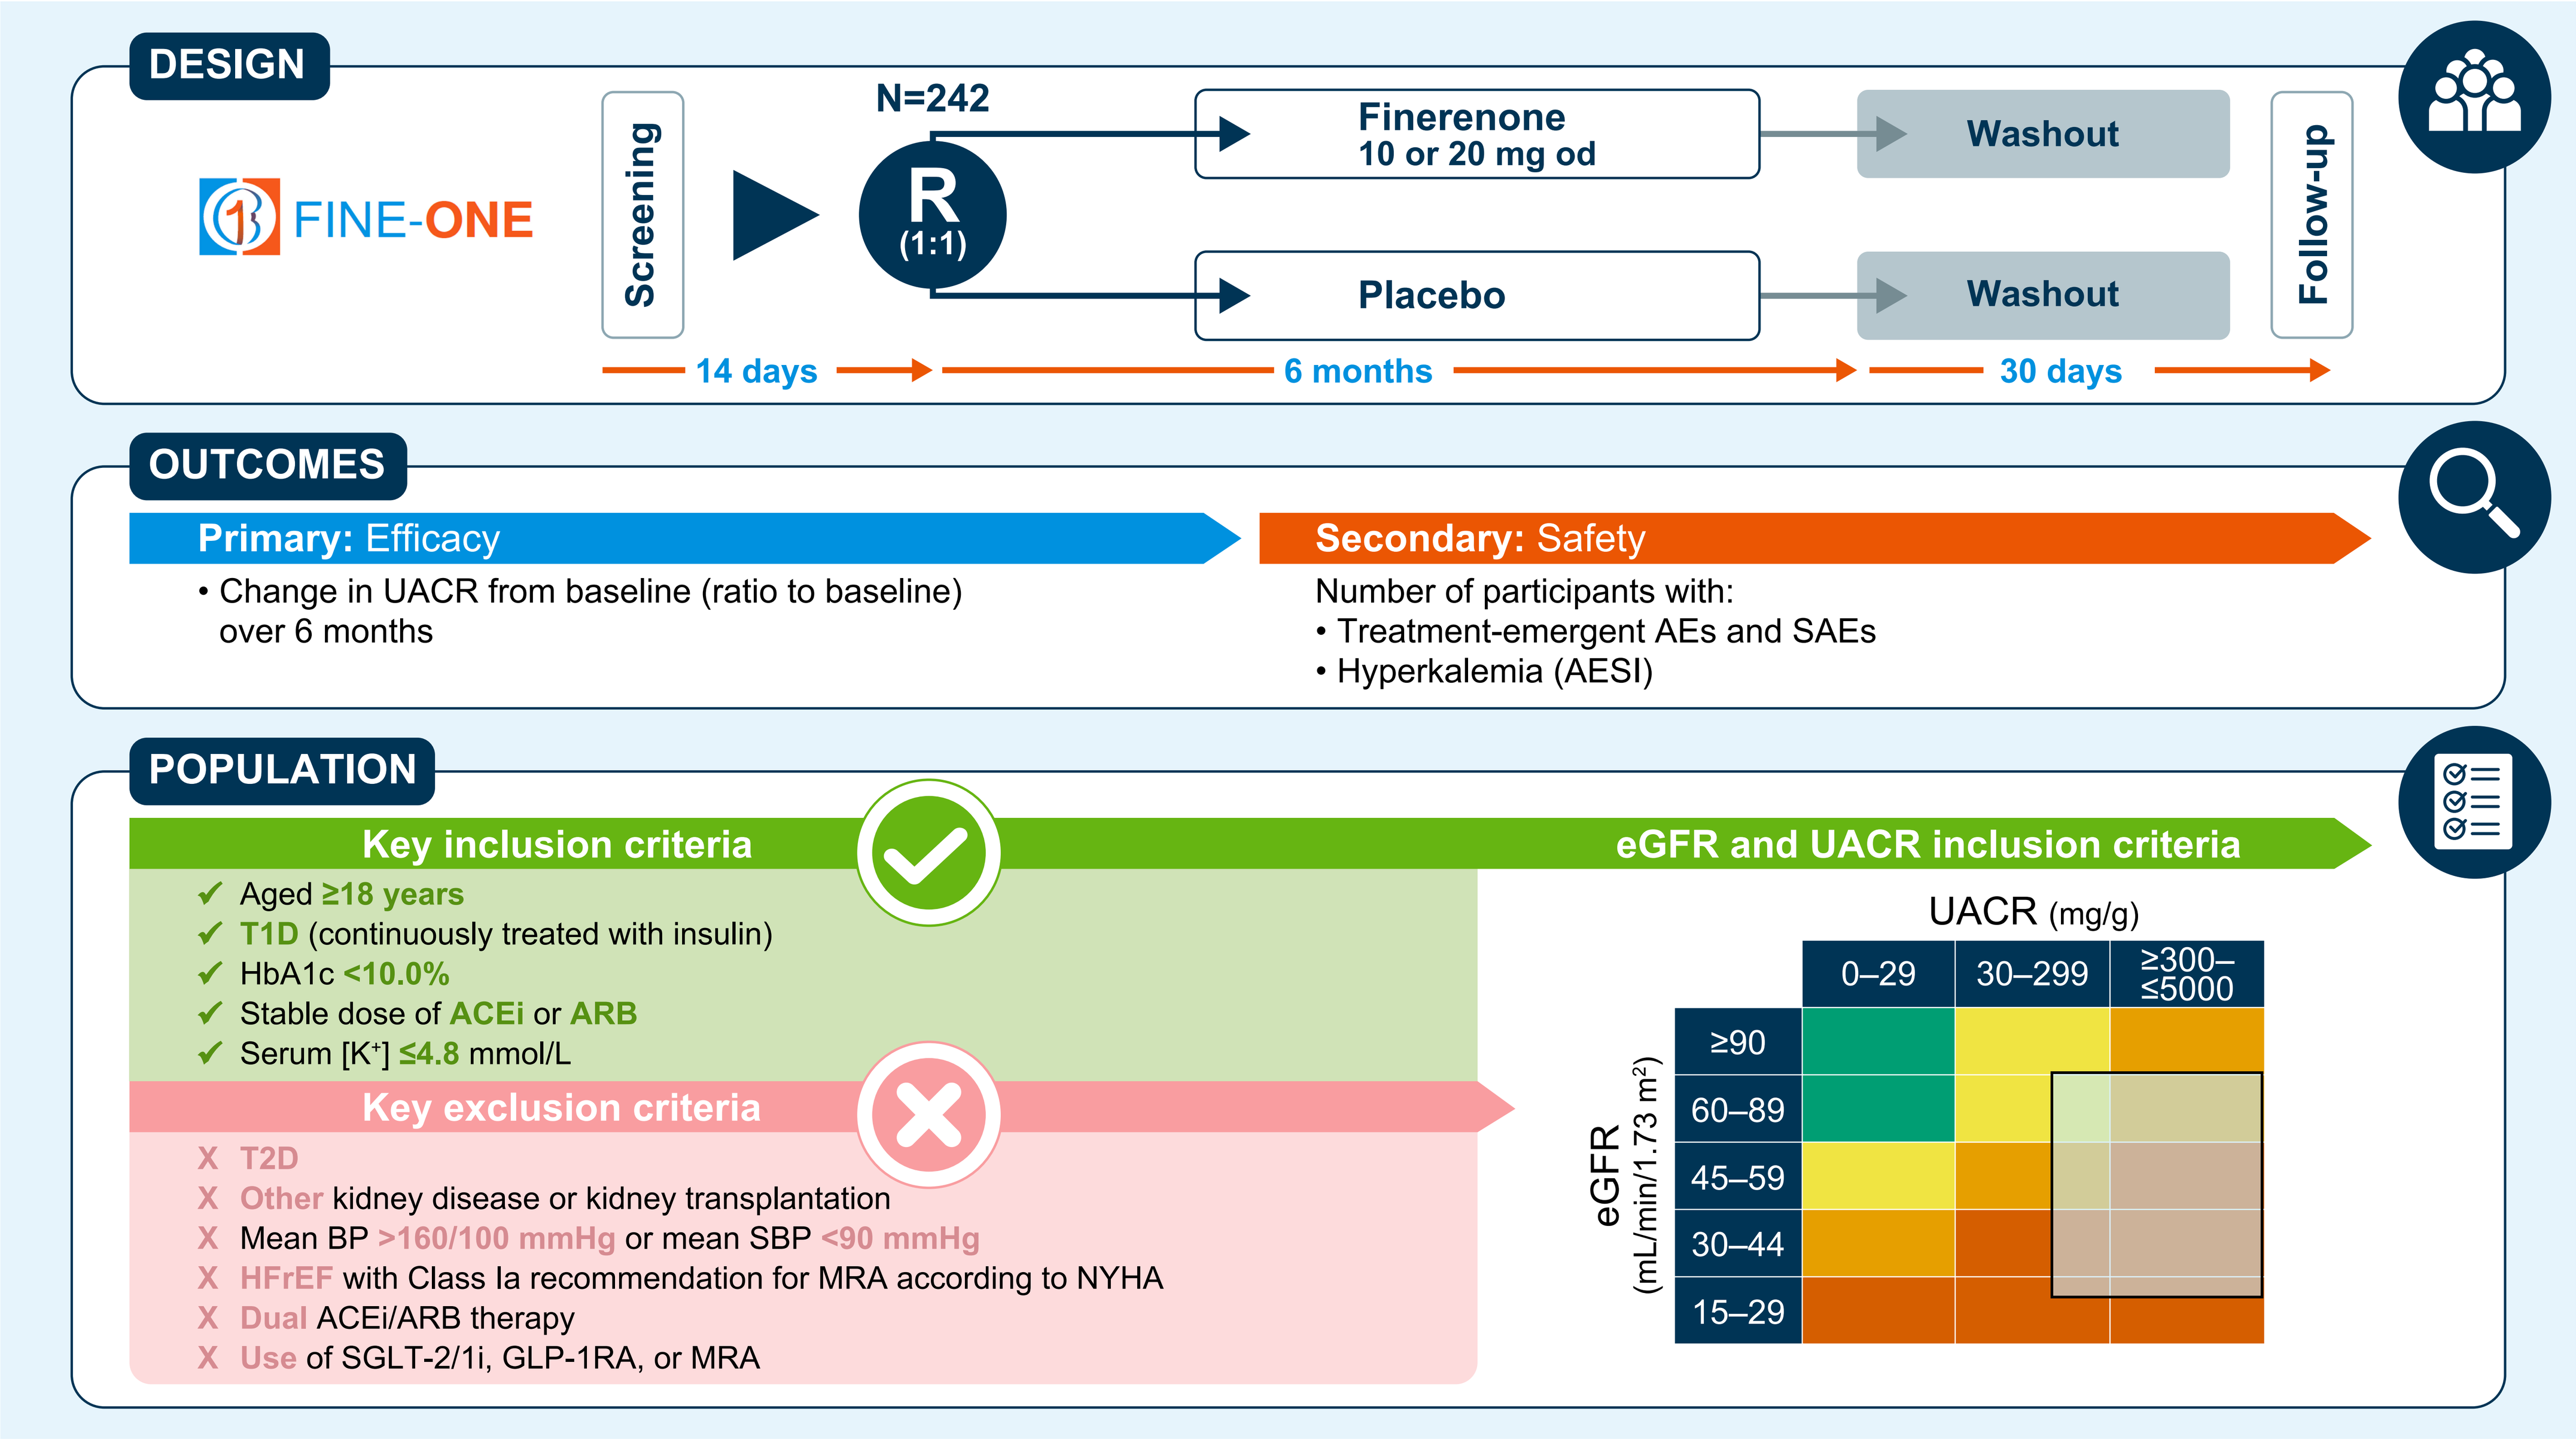
DESIGN
FINE-ONE
Screening
N=242
R
(1:1)
Finerenone
10 or 20 mg od
Placebo
Washout
Washout
Follow-up
14 days
6 months
30 days
OUTCOMES
Primary: Efficacy
• Change in UACR from baseline (ratio to baseline) over 6 months
Secondary: Safety
Number of participants with:
• Treatment-emergent AEs and SAEs
• Hyperkalemia (AESI)
POPULATION
Key inclusion criteria
✓ Aged ≥18 years
✓ T1D (continuously treated with insulin)
✓ HbA1c <10.0%
✓ Stable dose of ACEi or ARB
✓ Serum [K+] ≤4.8 mmol/L
Key exclusion criteria
X T2D
X Other kidney disease or kidney transplantation
X Mean BP >160/100 mmHg or mean SBP <90 mmHg
X HFrEF with Class Ia recommendation for MRA according to NYHA
X Dual ACEi/ARB therapy
X Use of SGLT-2/1i, GLP-1RA, or MRA
eGFR and UACR inclusion criteria
UACR (mg/g)
0–29
30–299
≥300–≤5000
eGFR (mL/min/1.73 m2)
≥90
60–89
45–59
30–44
15–29

AE, adverse event; ACEi, angiotensin-converting enzyme inhibitor; AESI, adverse event of special interest; ARB, angiotensin receptor blocker; BP, blood pressure; eGFR estimated glomerular filtration rate; GLP-1RA, glucagon-like peptide-1 receptor agonists; HbA1c, glycated hemoglobin; HFrEF, heart failure with reduced ejection fraction; $[K^+]$, potassium concentration; MRA, mineralocorticoid receptor antagonist; NYHA, New York Heart Association; R, randomization; SAE, serious adverse event; SBP, systolic blood pressure; SGLT-2/1i, sodium-glucose co-transporter-2/1 inhibitor; T1D, type 1 diabetes; T2D, type 2 diabetes; UACR, urine albumin-to-creatinine ratio

- Demographic characteristics, medical history, blood pressure, weight, and concomitant medications were documented for participants at the screening visit
 - Demographic characteristics included age, sex, race, ethnicity, and geographic region
- Blood and urine samples were collected for assessment of UACR, eGFR, HbA1c, and serum $[K^+]$
- Descriptive summary statistics including mean and standard deviation (SD), median and first and third quartiles (Q1, Q3), or n (%) were used to describe baseline characteristics

Results

Demographics

- FINE-ONE randomized 242 participants with CKD and T1D from Europe, North America, and Asia (**Table 1**)
 - Most participants were male (65%), and the mean age was 51.6 years
 - The most common race was White (72%), followed by Asian (20%) and Black (6%)

Table 1. Baseline demographic and clinical characteristics in FINE-ONE

Characteristic	Total N=242
Age, years, mean ± SD	51.6 ± 13.7
Sex, male, n (%)	158 (65.3)
Race, n (%)	
White	173 (71.5)
Asian	48 (19.8)
Black/African American	15 (6.2)
Other*	6 (2.5)
Region, n (%)	
Europe	120 (49.6)
North America	83 (34.3)
Asia	39 (16.1)
Weight, kg, mean ± SD	80.7 ± 20.8
BMI, kg/m^2, mean ± SD	27.6 ± 6.0
Blood pressure, mmHg, mean ± SD	
Systolic	135.2 ± 16.7
Diastolic	77.5 ± 10.8
Medical history	
Duration of diabetes, years, mean ± SD	32.0 ± 14.2
History of CVD,# n (%)	59 (24.4)
History of hypertension, n (%)	207 (85.5)
Medication use,‡ n (%)	
ACEi	112 (46.3)
ARBs	128 (52.9)
Diuretics	87 (36.0)
Beta blockers	66 (27.3)
Statins	178 (73.6)

*American Indian or Alaska Native, Native Hawaiian or Other Pacific Islander, or not reported; #history of CVD was determined by the presence of one of the following medical history preferred terms: myocardial infarction, coronary artery stenosis, cerebrovascular accident, transient ischemic attack, peripheral arterial occlusive disease, or cardiac failure; ‡use of SGLT-2i and GLP-1RA was not permitted in the FINE-ONE trial

ACEi, angiotensin-converting enzyme inhibitor; ARB, angiotensin receptor blocker; BMI, body mass index; CVD, cardiovascular disease; GLP-1RA, glucagon-like peptide-1 receptor agonist; SD, standard deviation; SGLT-2i, sodium-glucose co-transporter-2 inhibitor

Medical history and medication use

- A high proportion of participants had a history of hypertension (86%) (**Table 1**)
 - Mean blood pressure at baseline was 135.2/77.5 mmHg
 - In addition to ACEi or ARB therapy in >99% of participants, 36% of participants were on diuretics and 27% were on beta blockers
- Approximately a quarter of participants had a history of cardiovascular disease
 - Statins were used by 74% of participants
- The mean duration of diabetes at baseline was 32.0 years

Laboratory data

- At baseline, participants had a median UACR of 549 mg/g (interquartile range, 299–1191 mg/g) and 74% had a UACR of ≥300 mg/g (**Figure 2**)
 - Mean eGFR at baseline was 58.8 mL/min/1.73 m^2, and 48% of participants had a value ≥60 mL/min/1.73 m^2 with existing albuminuria
 - Mean serum $[K^+]$ was 4.6 mmol/L
- Participants had a mean HbA1c of 7.6%

Figure 2. FINE-ONE baseline laboratory data

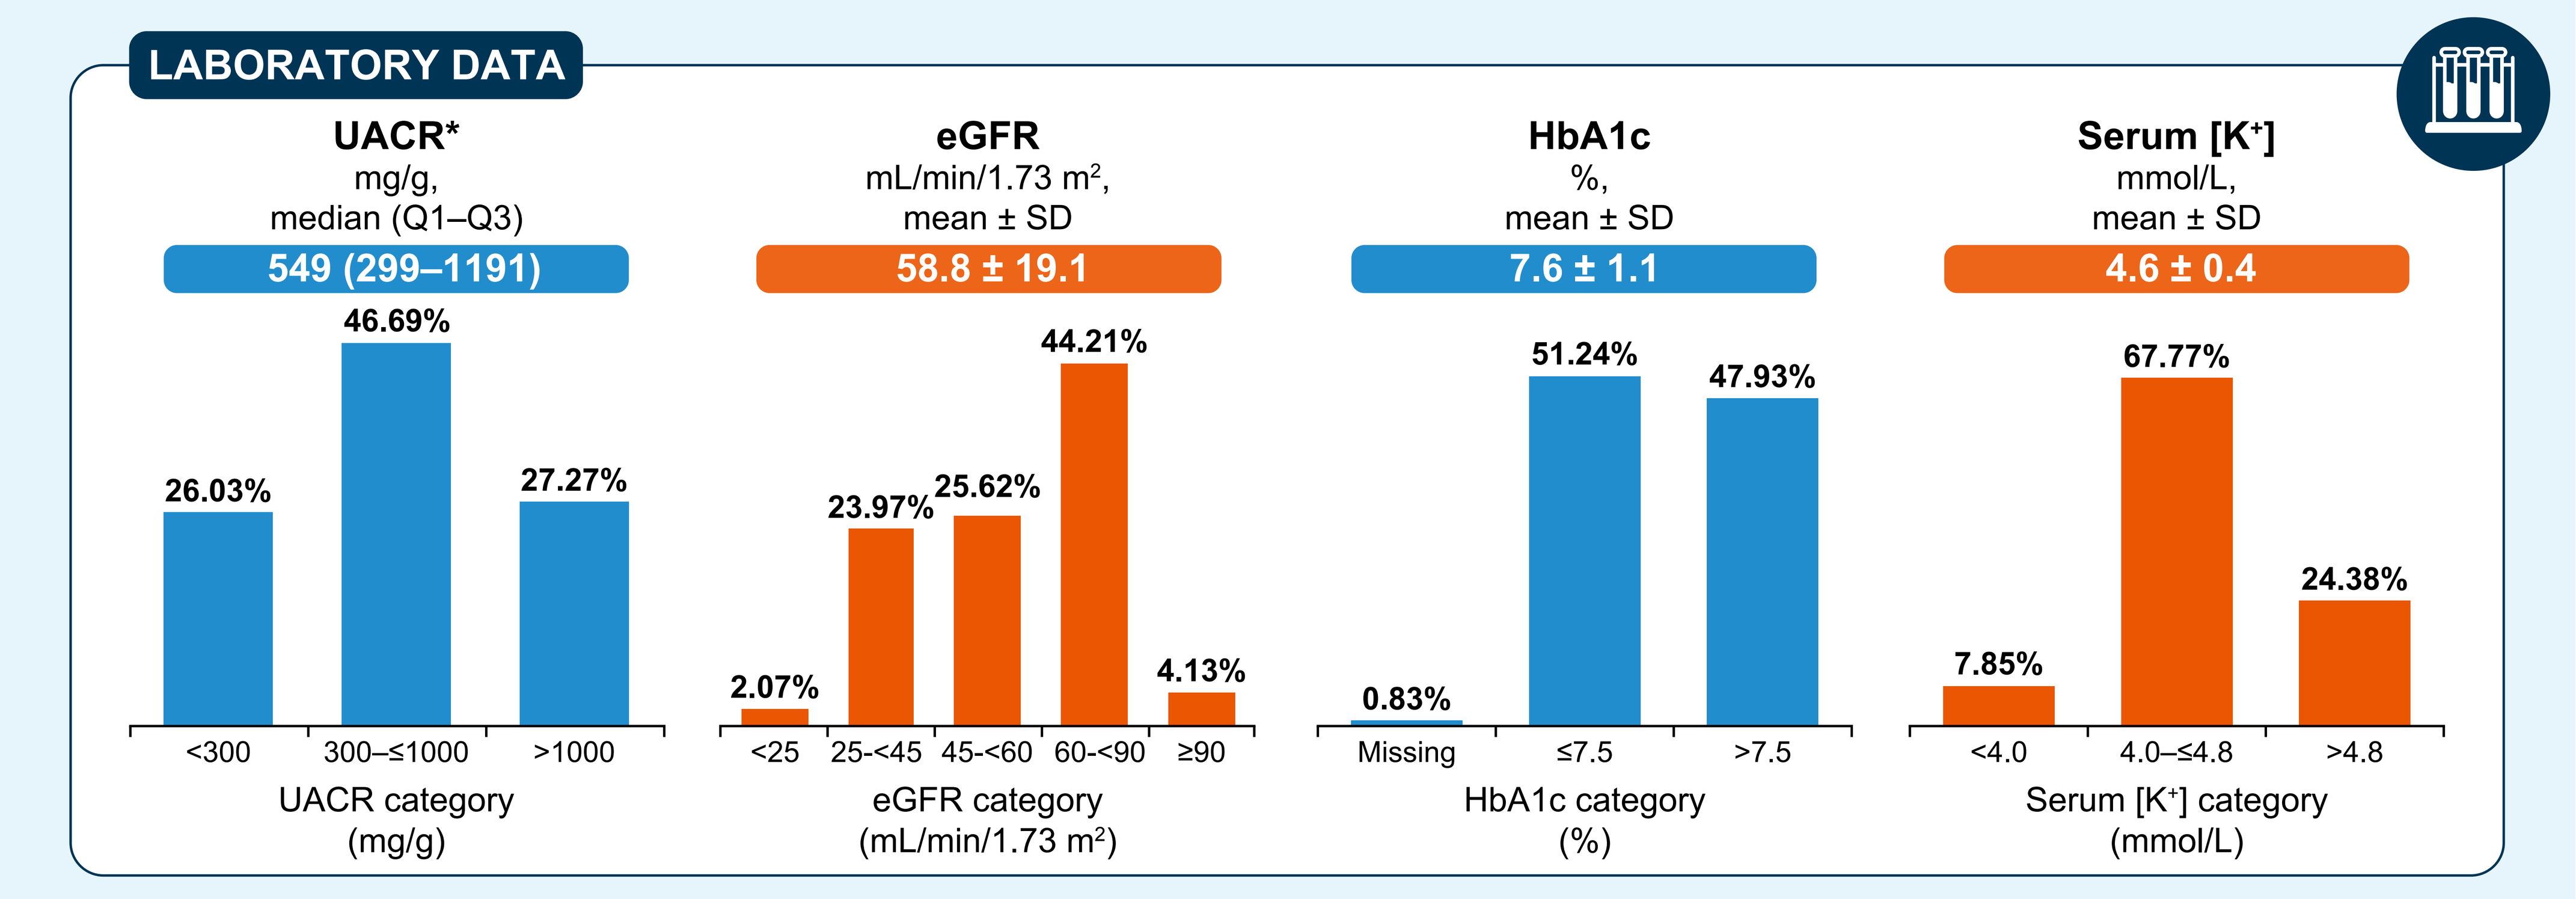
LABORATORY DATA
UACR*
mg/g,
median (Q1–Q3)
549 (299–1191)
26.03%
46.69%
27.27%
<300
300–≤1000
>1000
UACR category
(mg/g)
eGFR
mL/min/1.73 m2,
mean ± SD
58.8 ± 19.1
2.07%
23.97%
25.62%
44.21%
4.13%
<25
25-<45
45-<60
60-<90
≥90
eGFR category
(mL/min/1.73 m2)
HbA1c
%,
mean ± SD
7.6 ± 1.1
0.83%
51.24%
47.93%
Missing
≤7.5
>7.5
HbA1c category
(%)
Serum [K+]
mmol/L,
mean ± SD
4.6 ± 0.4
7.85%
67.77%
24.38%
<4.0
4.0–≤4.8
>4.8
Serum [K+] category
(mmol/L)

*Proportions may not add up to 100% due to rounding

eGFR estimated glomerular filtration rate; HbA1c, glycated hemoglobin; $[K^+]$, potassium concentration; Q, quartile; SD; standard deviation; UACR, urine albumin-to-creatinine ratio

Conclusions

- FINE-ONE was a global trial that randomized a high-risk population with CKD and T1D, characterized by wide ranges of both albuminuria and eGFR
- Participants had high rates of hypertension and high risks of progressive loss of kidney function
- Determining the effect of finerenone on albuminuria in a representative population with CKD and T1D could help translate the findings from people with CKD and T2D in FIDELITY to those with CKD and T1D in FINE-ONE

References

1. Tuttle K, *et al. Lancet Reg Health Am* 2025;47:101130.
2. de Boer IH, *et al. Diabetes Care* 2022;45:3075–3090.
3. American Diabetes Association Professional Practice Committee. *Diabetes Care* 2025; 48:S239–s251.
4. Colombo M, *et al. Diabetologia* 2020;63:636–647.
5. Heerspink HJL, *et al. Diabetes Res Clin Pract* 2023;204:110908.
6. Agarwal R, *et al. Eur Heart J* 2022;43:474–484.
7. Pitt B, *et al. N Engl J Med* 2021;385:2252–2263.
8. Bakris GL, *et al. N Engl J Med* 2020;383:2219–2229.

Acknowledgments

The FINE-ONE study was funded by Bayer AG. Medical writing and editorial support were provided by Alison McTavish, MSc (HCG), with funding from Bayer AG.

Poster 875-P presented at the 85th Scientific Sessions of the American Diabetes Association (ADA), Chicago, IL, June 20–23, 2025